A phase 1/2 study of the next-generation Nectin-4-targeting antibody–drug conjugate CRB-701 (SYS6002) in patients with recurrent or metastatic head and neck squamous cell carcinoma

Charlene Mantia[1]*, Glenn J Hanna[1]*, Yohann Loriot[2], Peter Vu[3], Diego Tosi[4], Tudor-Eliade Ciuleanu[5], Abhishek Tripathi[6], Pamela Munster[7], Aarti Bhatia[8], Lauren Shih[9], Andrea Zivi[10], Ik Shin Chin[11], Maria Julia Lostes[12], Frédéric Rolland[13], Ivan Barrera[14], Kurt Preugschat[14], Paola M Grant[15], Ian Hodgson[15], Cesar A Perez[16], Ari J Rosenberg[17]

[1]Dana-Farber Cancer Institute, Harvard Medical School, Boston, MA, USA; [2]Institut Gustave Roussy, Université Paris-Saclay, Villejuif, France; [3]University of California San Diego, San Diego, CA, USA; [4]Institut Régional du Cancer de Montpellier-Val d'Aurelle, Montpellier, France; [5]Arensia Research Clinic, Cluj-Napoca, Romania; [6]City of Hope, Duarte, CA, USA; [7]University of California San Francisco, San Francisco, CA, USA; [8]Yale School of Medicine, Yale Cancer Center, New Haven, CT, USA; [9]Fred Hutchinson Cancer Center, University of Washington, Seattle, WA, USA; [10]Azienda Ospedaliera Universitaria Integrata di Verona, Centro Ricerche Cliniche di Verona, Verona, Italy; [11]Clatterbridge Cancer Centre Liverpool, The Clatterbridge Cancer Centre NHS Foundation Trust, Liverpool, UK; [12]Hospital Universitari Vall d'Hebron, Barcelona, Spain; [13]Institut de Cancérologie de l'Ouest, Saint Herblain, France; [14]Precision for Medicine, Flemington, NJ, USA; [15]Corbus Pharmaceuticals, Inc., Norwood, MA, USA; [16]Sarah Cannon Research Institute at Florida Cancer Specialists, Orlando, FL, USA; [17]University of Chicago, Chicago, IL, USA

*Both authors contributed equally

Abstract ID: A6062
Poster board: 519

Scan QR code to download a copy of this poster.
Copies of this poster obtained through the QR code are for personal use only and may not be reproduced without permission from ASCO® or the authors of this poster.

Correspondence: cesar.perezbatista@flcancer.com
arirosenberg@bsd.uchicago.edu

BACKGROUND

- Head and neck squamous cell carcinoma (HNSCC) is the seventh most common malignancy worldwide, accounting for approximately 4.5% of all cancers globally.[1]
 - Oropharyngeal squamous cell carcinoma (OPSCC) accounts for approximately 31% of HNSCC cases in the USA, with approximately 70% of cases attributable to human papillomavirus (HPV) infection.[2,3]
- Nectin-4 (*PVRL4*) expression has been detected in approximately 86% of HNSCC tumors, with higher levels in HPV-associated diseases.[4]
 - HPV-positive tumors often demonstrate an inflamed tumor microenvironment with increased immune infiltration and elevated programmed death-ligand 1 (PD-L1) expression.[5–7]
- CRB-701 is a next-generation Nectin-4-targeted monomethyl auristatin E (MMAE)-based antibody–drug conjugate (ADC; **Figure 1**).
- Previously reported results from the ongoing phase 1/2 CRB-701-01 study (NCT06265727) demonstrated a manageable safety profile and encouraging efficacy signals in heavily pretreated patients with recurrent or metastatic (R/M) HNSCC treated with CRB-701.[8]
- This poster presents updated efficacy data from an additional 30 patients (total N = 71) treated with CRB-701 2.7 mg/kg or 3.6 mg/kg Q3W, including 6 months of extended follow-up.

Figure 1. CRB-701

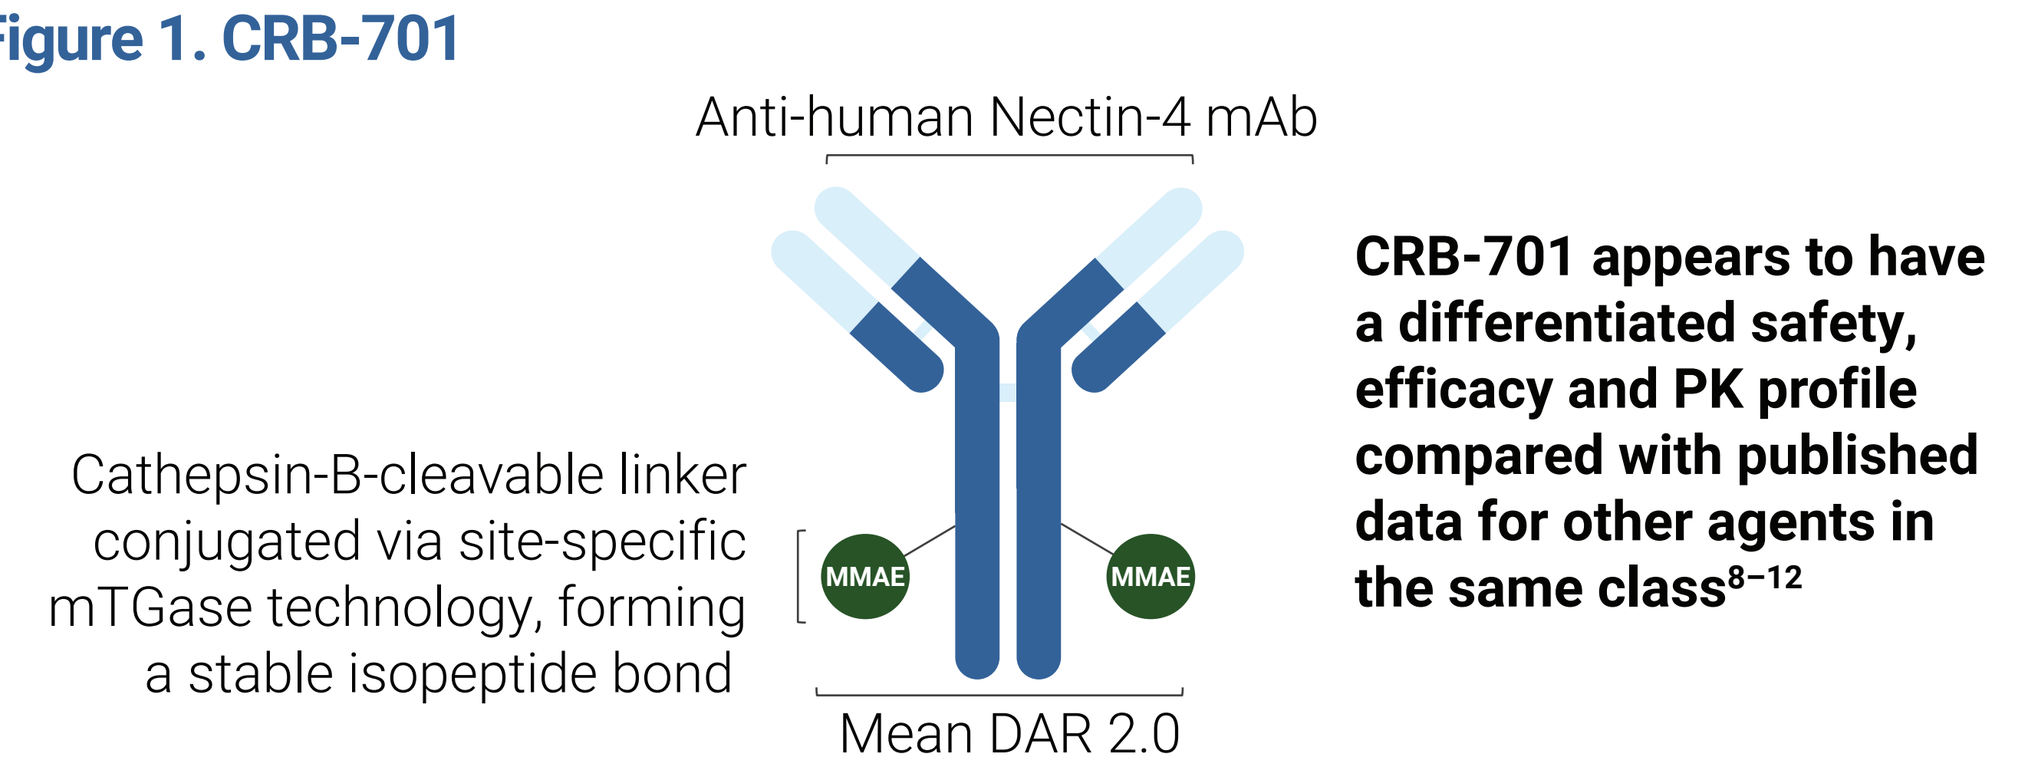
Anti-human Nectin-4 mAb
Cathepsin-B-cleavable linker conjugated via site-specific mTGase technology, forming a stable isopeptide bond
MMAE
MMAE
Mean DAR 2.0
CRB-701 appears to have a differentiated safety, efficacy and PK profile compared with published data for other agents in the same class[8–12]

DAR, drug:antibody ratio; mAb, monoclonal antibody; MMAE, monomethyl auristatin E; mTGase, microbial transglutaminase; PK, pharmacokinetics.

METHODS

- The study design is presented in **Figure 2**.
- Tumor type and subtype were confirmed from previous diagnostic records based on histology.

Figure 2. Study design

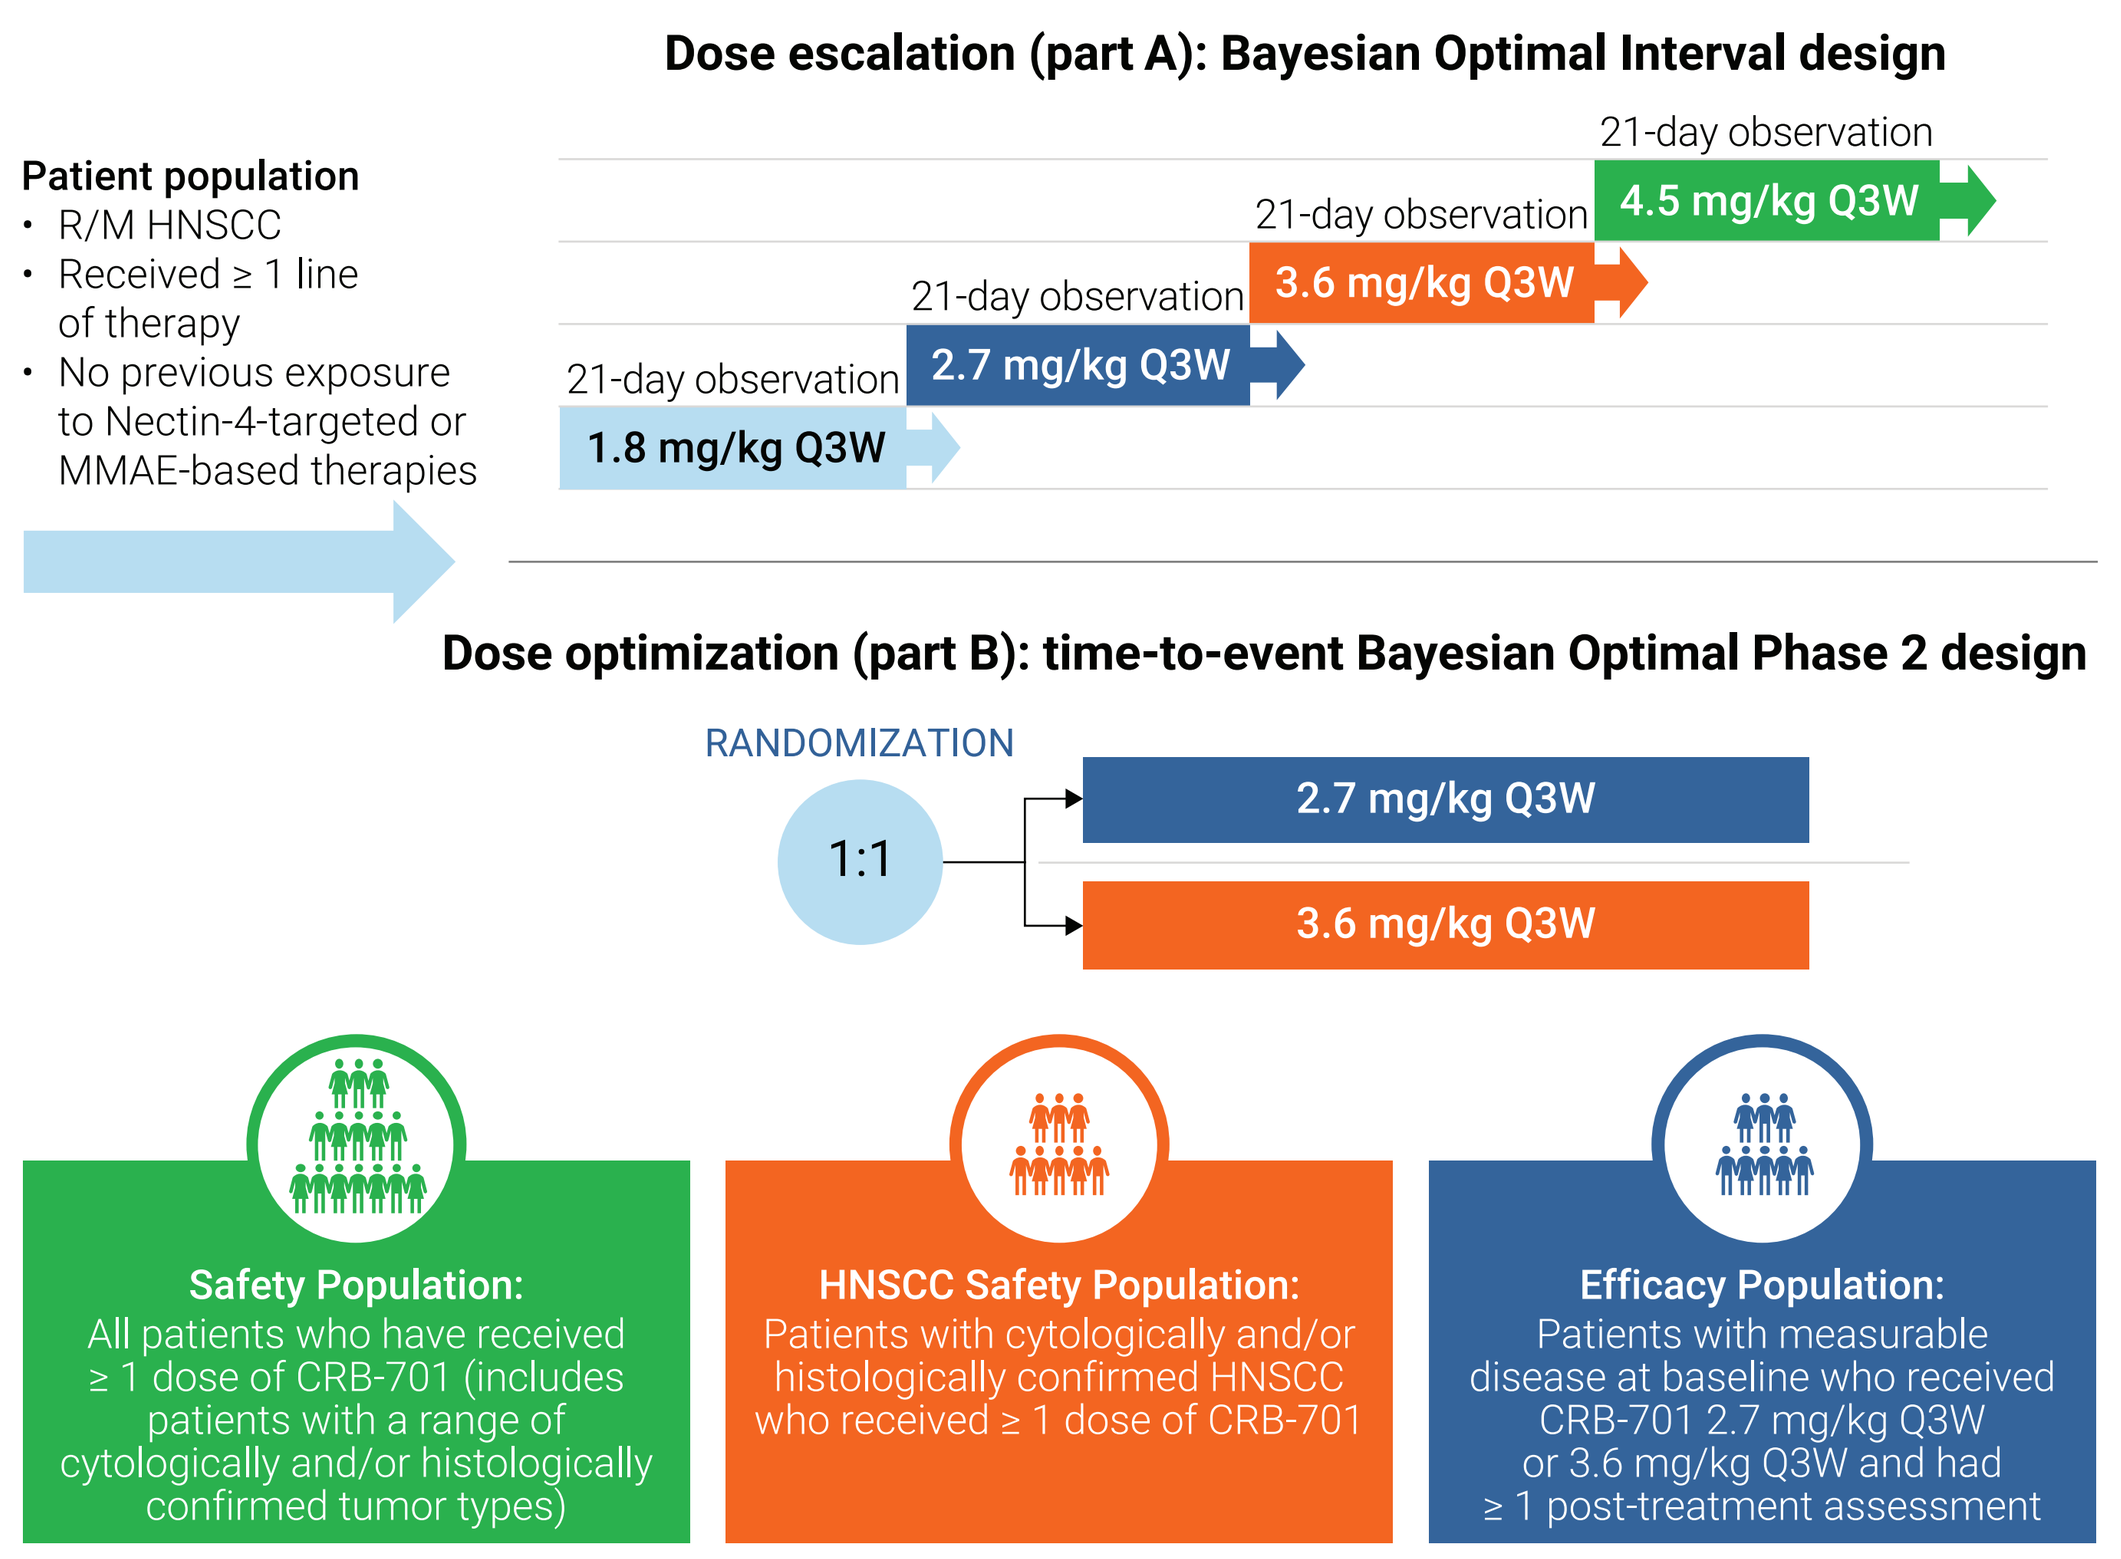
Dose escalation (part A): Bayesian Optimal Interval design
Patient population
• R/M HNSCC
• Received ≥ 1 line of therapy
• No previous exposure to Nectin-4-targeted or MMAE-based therapies
21-day observation
4.5 mg/kg Q3W
21-day observation
3.6 mg/kg Q3W
21-day observation
2.7 mg/kg Q3W
21-day observation
1.8 mg/kg Q3W
Dose optimization (part B): time-to-event Bayesian Optimal Phase 2 design
RANDOMIZATION
1:1
2.7 mg/kg Q3W
3.6 mg/kg Q3W
Safety Population:
All patients who have received ≥ 1 dose of CRB-701 (includes patients with a range of cytologically and/or histologically confirmed tumor types)
HNSCC Safety Population:
Patients with cytologically and/or histologically confirmed HNSCC who received ≥ 1 dose of CRB-701
Efficacy Population:
Patients with measurable disease at baseline who received CRB-701 2.7 mg/kg Q3W or 3.6 mg/kg Q3W and had ≥ 1 post-treatment assessment

In part A, dose escalation/de-escalation decisions were made based on the occurrence of dose-limiting toxicities. In part B, dose levels were defined by the pharmacologically active dose range identified in part A.
HNSCC, head and neck squamous cell carcinoma; MMAE, monomethyl auristatin E; Q3W, every 3 weeks; R/M, recurrent or metastatic.

- HPV status was retrospectively determined using p16 immunohistochemistry and/or HPV genomic assays.
- Confirmed objective response rate (cORR), duration of response (DoR) and progression-free survival (PFS) were assessed.
 - Responses were based on investigator assessment per Response Evaluation Criteria in Solid Tumours (RECIST) v1.1 criteria,[13] and a scan at least 4 weeks after the initial response was required to confirm partial and complete objective responses.
 - Subgroup analyses based on HNSCC subtype were also performed.

RESULTS

Patient disposition

- As of April 1, 2026, a total of 317 patients had received at least one dose of CRB-701 monotherapy. Among them, 75 patients with HNSCC were treated with 2.7 mg/kg or 3.6 mg/kg Q3W; 41 had OPSCC and 34 had non-oropharyngeal squamous cell carcinoma (non-OPSCC; **Table 1**).

Table 1. Study patient disposition

Disposition	Safety Population	Efficacy Population	Not evaluable[a]
Patients treated with CRB-701 monotherapy (all tumors and doses)	317	N/A	N/A
Patients with HNSCC treated with CRB-701 monotherapy (2.7 mg/kg or 3.6 mg/kg Q3W)	75	71	4
Patients with OPSCC treated with CRB-701 monotherapy (2.7 mg/kg or 3.6 mg/kg Q3W)	41	41	0
Patients with non-OPSCC treated with CRB-701 monotherapy (2.7 mg/kg or 3.6 mg/kg Q3W)	34	30	4

[a]Patients with no post-baseline scan assessment.
HNSCC, head and neck squamous cell carcinoma; N/A, not applicable; non-OPSCC, non-oropharyngeal squamous cell carcinoma; OPSCC, oropharyngeal squamous cell carcinoma.

Safety profile

- The most common treatment-related adverse events (TRAEs; occurring in ≥ 20% of the overall Safety Population) are presented in **Figure 3**.
 - The safety profile in patients with HNSCC was similar to that observed in the overall Safety Population.
- In patients with HNSCC, keratitis occurred more frequently in patients receiving CRB-701 3.6 mg/kg than 2.7 mg/kg (51.3% vs 36.1%; **Table 2**). Grade 3 events were also more common at the 3.6 mg/kg dose (12.8% vs 5.6%).
 - No grade 4 or 5 keratitis events were reported.
 - Dose interruptions and reductions were frequently implemented to manage ocular toxicity; however, discontinuations were uncommon.
 - Ocular toxicities were reversible and manageable with dose modifications and prophylactic eye drops.

Figure 3. Distribution of TRAEs reported in ≥ 20% of patients by maximum CTCAE grade (N = 317)

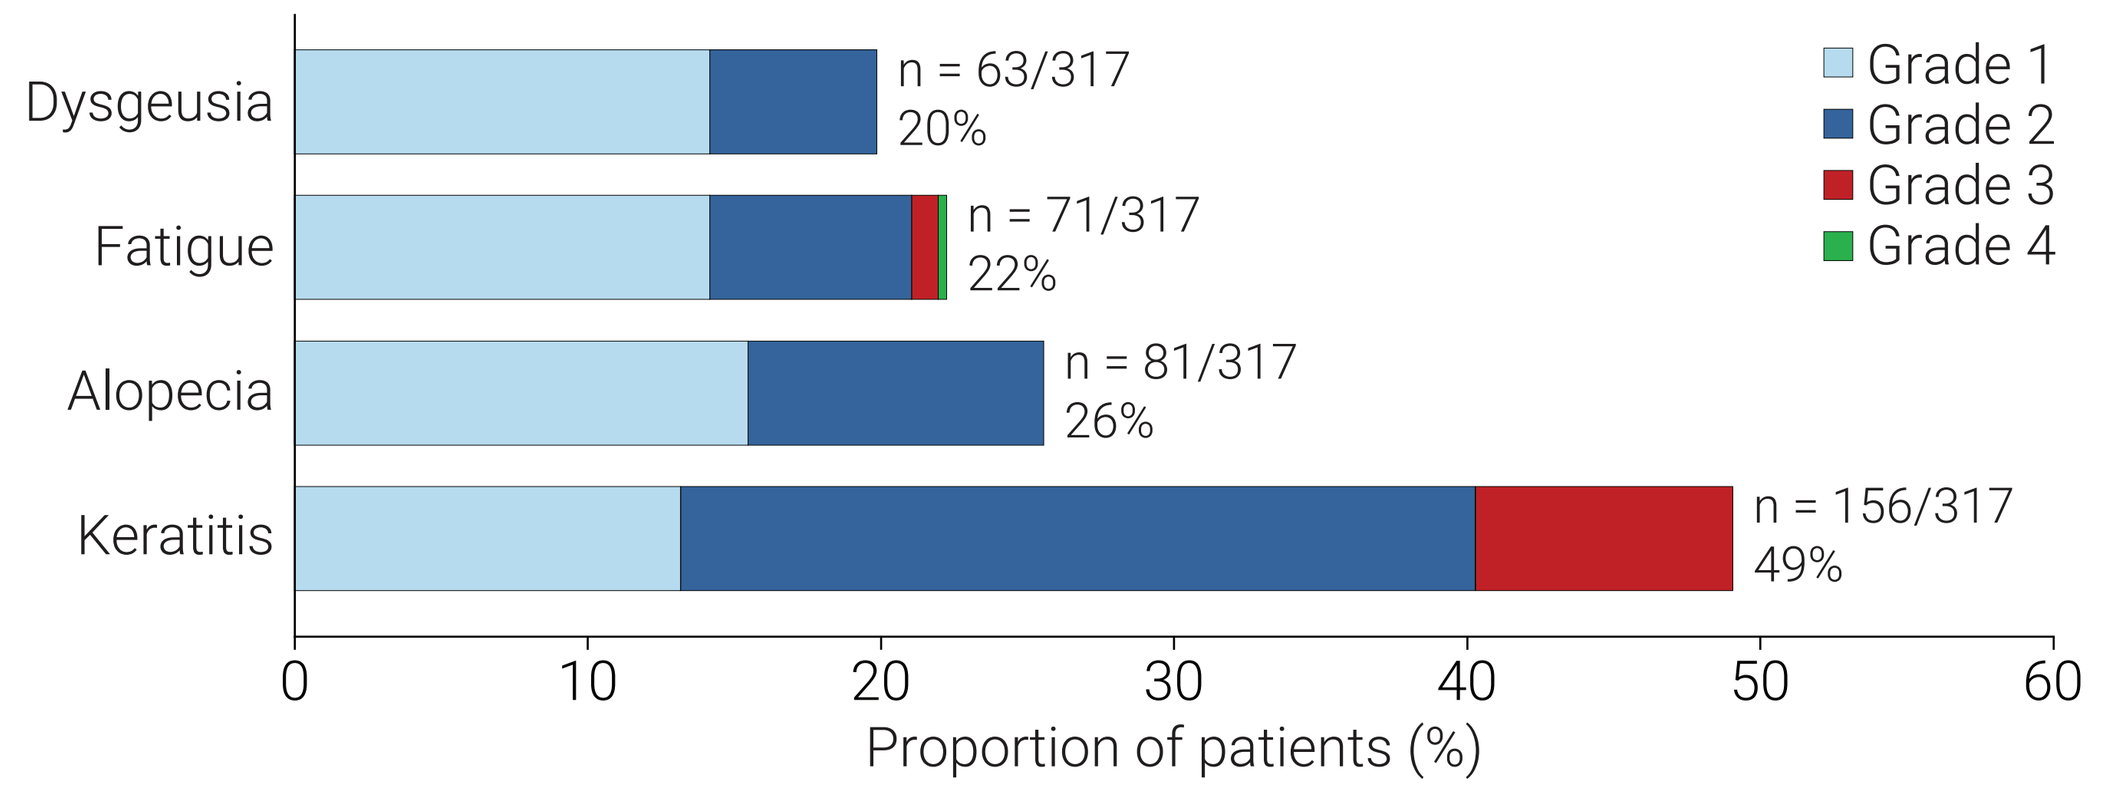
Dysgeusia
n = 63/317
20%
Fatigue
n = 71/317
22%
Alopecia
n = 81/317
26%
Keratitis
n = 156/317
49%
Grade 1
Grade 2
Grade 3
Grade 4
0
10
20
30
40
50
60
Proportion of patients (%)

Patients with multiple events of the same type are counted once at the highest grade.
CTCAE, Common Terminology Criteria for Adverse Events; TRAE, treatment-related adverse event.

Table 2. Ocular toxicity, extent of exposure and dose modifications in patients with HNSCC

Event	CRB-701 2.7 mg/kg Q3W (n = 36)	CRB-701 3.6 mg/kg Q3W (n = 39)	Overall (N = 75)
Keratitis, n (%)			
All grades	13 (36.1)	20 (51.3)	33 (44.0)
Grade ≥ 3	2 (5.6)	5 (12.8)	7 (9.3)
Extent of exposure			
Duration of exposure,[a] days, median (range)	86.0 (1–367)	93.0 (1–477)	87.0 (1–477)
Relative dose intensity,[b] %, median (range)	97.7 (56.1–103.3)	85.7 (38.0–101.6)	96.9 (38.0–103.3)
Dose modification due to ocular toxicity, n (%)			
Discontinuation	0	1 (2.6)	1 (1.3)
Reduction	7 (19.4)	14 (35.9)	21 (28.0)
Interruption or delay	13 (36.1)	18 (46.2)	31 (41.3)

[a]Duration of exposure to CRB-701 is calculated using the date of last dose of CRB-701 – the date of first dose of CRB-701 + 1.
[b]Relative dose intensity is defined as the ratio of the actual delivered dose to the planned dose, expressed as a percentage.
HNSCC, head and neck squamous cell carcinoma; Q3W, every 3 weeks.

Baseline characteristics of patients with HNSCC

- Key patient baseline characteristics for patients with HNSCC are presented in **Table 3**.

Table 3. Key patient baseline characteristics

Characteristic	OPSCC (n = 41)	non-OPSCC (n = 34)	HNSCC (N = 75)
Male, n (%)	37 (90.2)	30 (88.2)	67 (89.3)
Age, years, median (range)	62 (36–77)	62 (24–78)	62 (24–78)
ECOG PS, n (%)			
0	24 (58.5)	11 (32.4)	35 (46.7)
1	17 (41.5)	22 (64.7)	39 (52.0)
2	0	1 (2.9)	1 (1.3)
Number of previous therapies, median (range)	3 (1–9)	2 (1–7)	3 (1–9)
HPV status, n (%)			
Positive	35 (85.4)	8 (23.5)	43 (57.3)
Negative	6 (14.6)	24 (70.6)	30 (40.0)
Missing	0	2 (5.9)	2 (2.7)
Disease extent, n (%)			
Locally recurrent	2 (4.9)	10 (29.4)	12 (16.0)
Metastatic	39 (95.1)	24 (70.6)	63 (84.0)
Locally recurrent and metastatic	0	0	0

ECOG PS, Eastern Cooperative Oncology Group Performance Status; HNSCC, head and neck squamous cell carcinoma; HPV, human papillomavirus; non-OPSCC, non-oropharyngeal squamous cell carcinoma; OPSCC, oropharyngeal squamous cell carcinoma.

Efficacy in patients with HNSCC and by anatomical subtypes

- Of those in the Efficacy Population, 41 patients had OPSCC and 30 patients had non-OPSCC.
- Distinct efficacy outcomes were observed between HNSCC subtypes, with superior confirmed antitumor responses (cORR), DoR and PFS noted in the OPSCC population at the 3.6 mg/kg Q3W dose (**Figure 4** and **Table 4**).
 - The majority of patients with OPSCC were HPV-positive and had been previously exposed to both platinum-based chemotherapy and anti-PD-L1 therapy.
 - Most clinical responses were observed in patients with HPV-positive OPSCC.
 - Antitumor responses were observed in patients with high and low levels of Nectin-4 expression (data not shown).
 - Antitumor responses were observed across all subgroups when stratified by prior lines of therapy (≤ 3 vs > 3) or disease extent (locally recurrent vs metastatic; data not shown).

Figure 4. Percentage change from baseline in sum of diameters with confirmed best overall response (A) and patient-level best overall response by dose (B) for CRB-701 Q3W

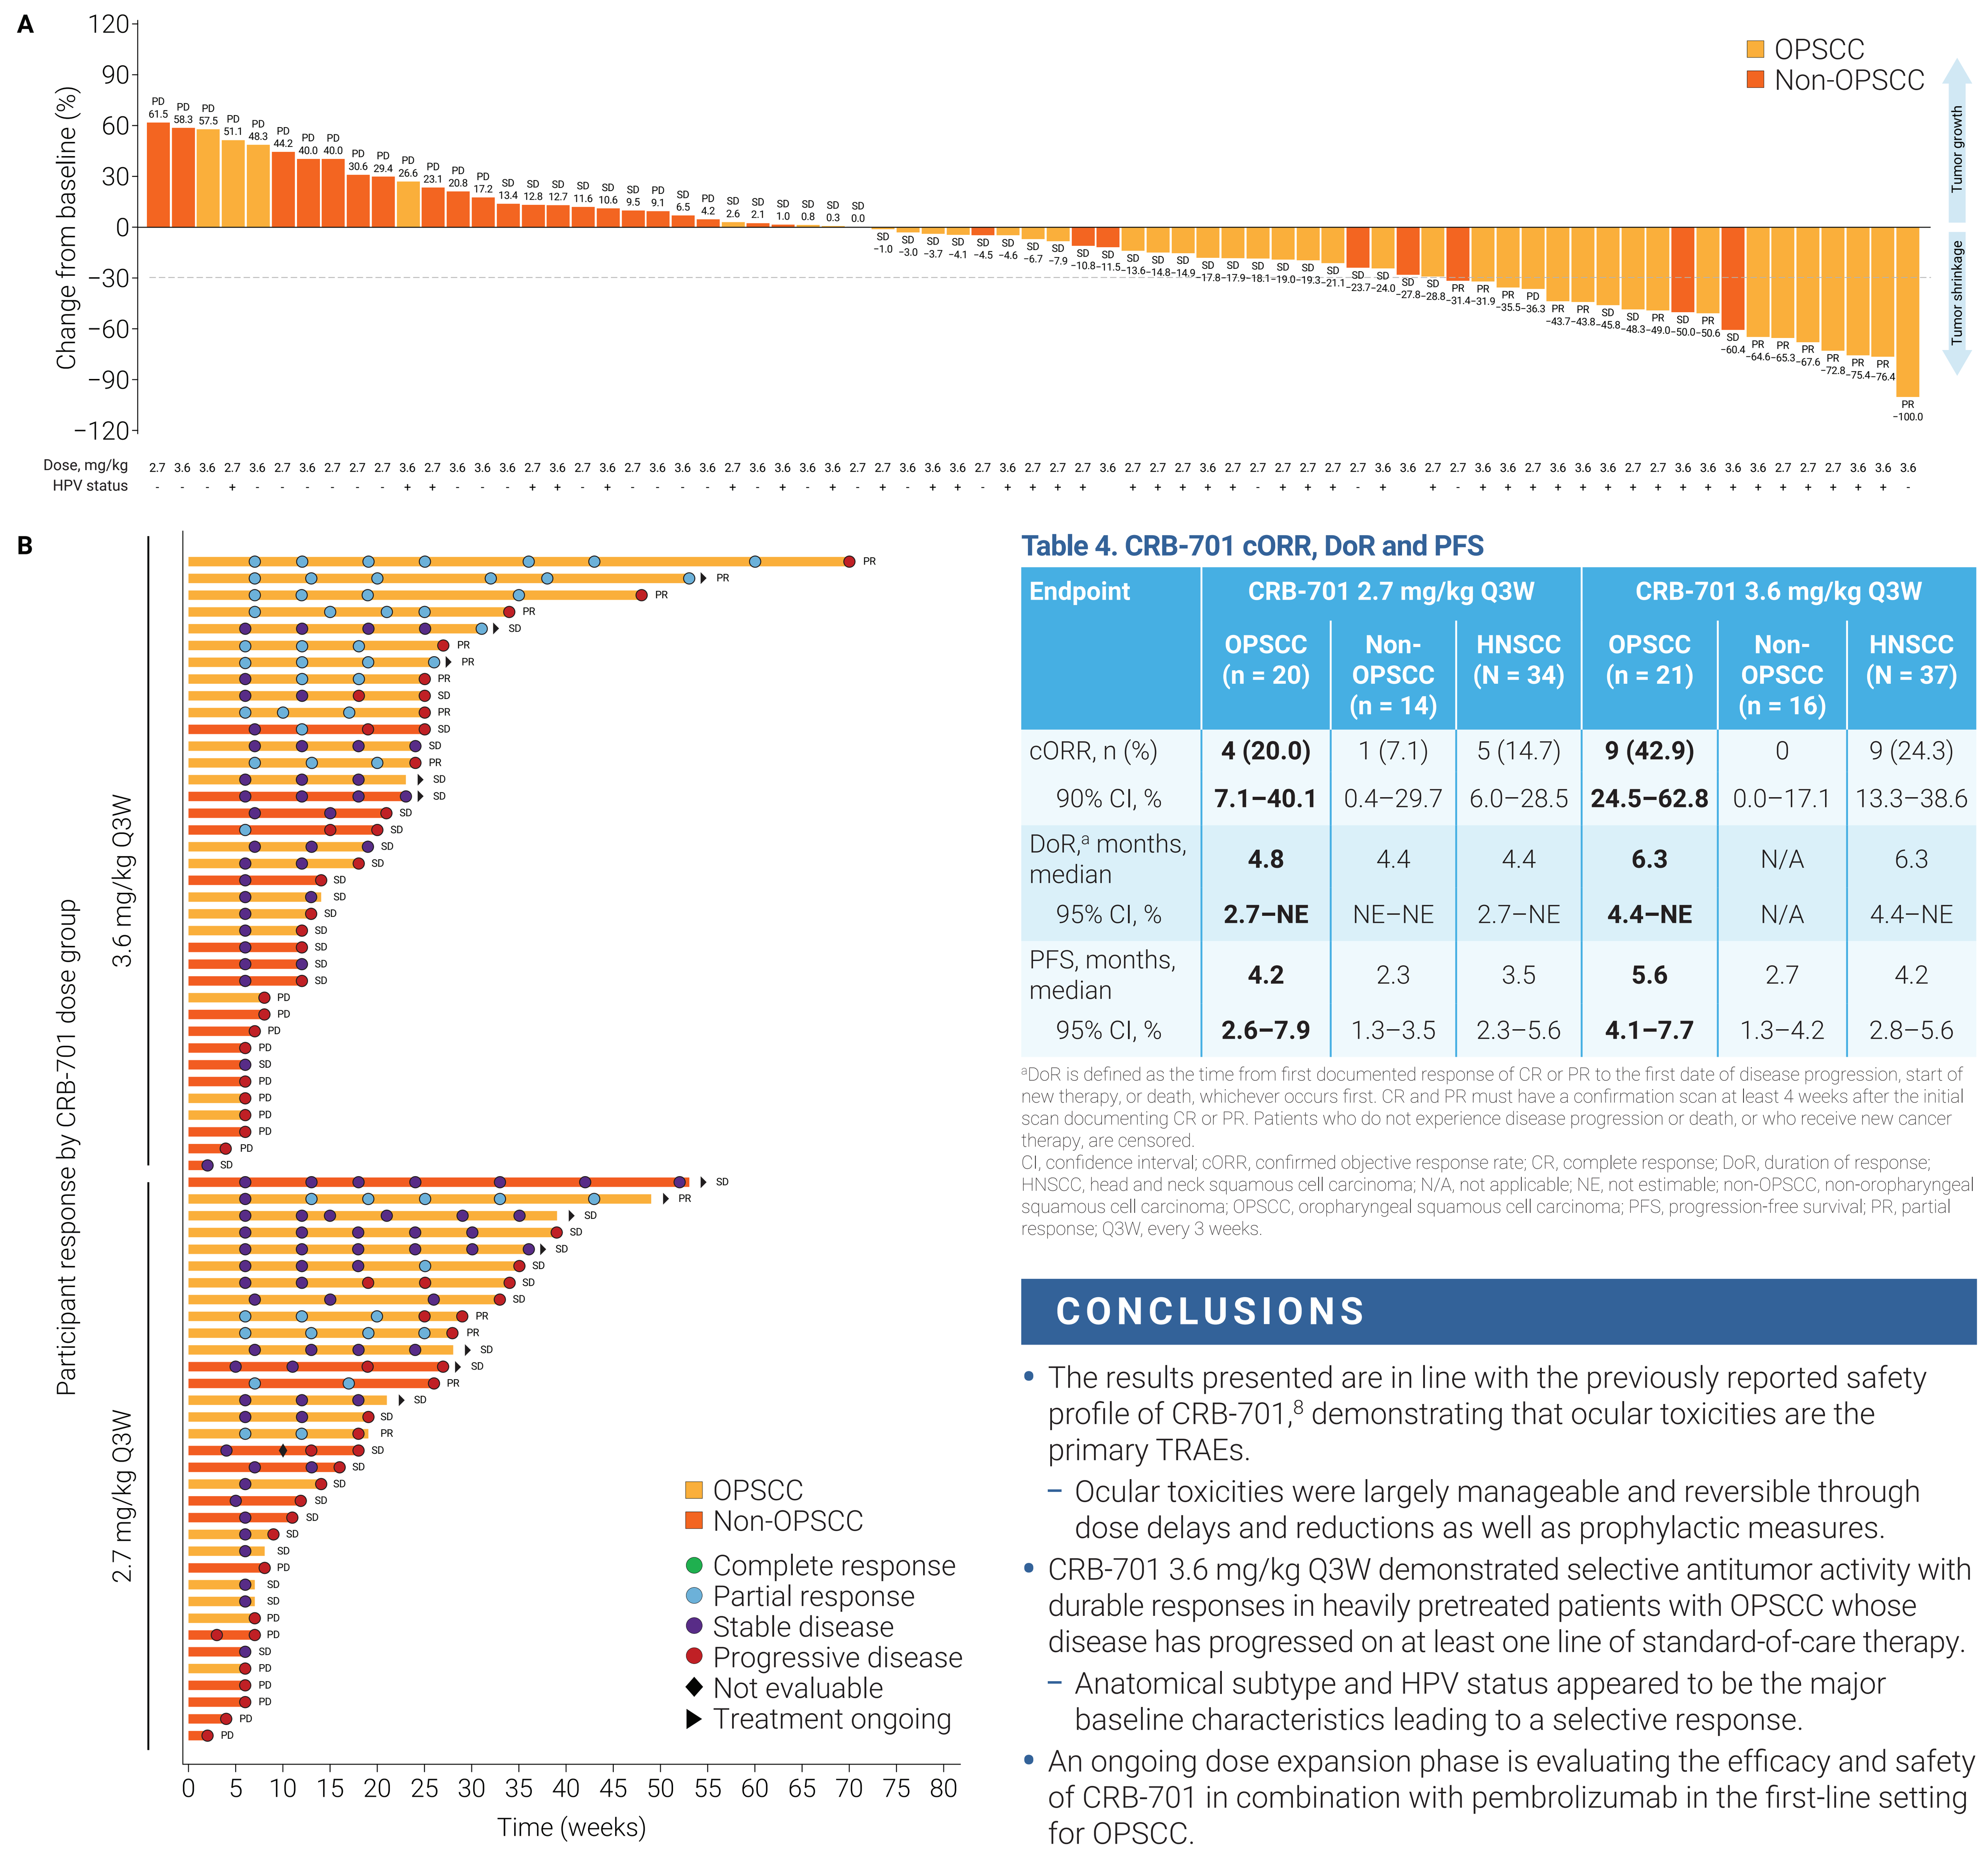
A
Change from baseline (%)
OPSCC
Non-OPSCC
Tumor growth
Tumor shrinkage
Dose, mg/kg
HPV status
B
Participant response by CRB-701 dose group
3.6 mg/kg Q3W
2.7 mg/kg Q3W
OPSCC
Non-OPSCC
Complete response
Partial response
Stable disease
Progressive disease
Not evaluable
Treatment ongoing
Time (weeks)

Data are summarized based on the dose group assigned at enrollment. Each bar represents one participant. Best overall response is indicated at the end of each bar. Dashed line on panel A indicates the threshold for PR according to RECIST v1.1 criteria[13] (> 30% reduction in the sum of diameters).
CR, complete response; non-OPSCC, non-oropharyngeal squamous cell carcinoma; OPSCC, oropharyngeal squamous cell carcinoma; PD, progressive disease; PR, partial response; Q3W, every 3 weeks; RECIST, Response Evaluation Criteria in Solid Tumours; SD, stable disease.

Table 4. CRB-701 cORR, DoR and PFS

Endpoint	CRB-701 2.7 mg/kg Q3W			CRB-701 3.6 mg/kg Q3W		
	OPSCC (n = 20)	Non-OPSCC (n = 14)	HNSCC (N = 34)	OPSCC (n = 21)	Non-OPSCC (n = 16)	HNSCC (N = 37)
cORR, n (%)	**4 (20.0)**	1 (7.1)	5 (14.7)	**9 (42.9)**	0	9 (24.3)
90% CI, %	**7.1–40.1**	0.4–29.7	6.0–28.5	**24.5–62.8**	0.0–17.1	13.3–38.6
DoR,[a] months, median	**4.8**	4.4	4.4	**6.3**	N/A	6.3
95% CI, %	**2.7–NE**	NE–NE	2.7–NE	**4.4–NE**	N/A	4.4–NE
PFS, months, median	**4.2**	2.3	3.5	**5.6**	2.7	4.2
95% CI, %	**2.6–7.9**	1.3–3.5	2.3–5.6	**4.1–7.7**	1.3–4.2	2.8–5.6

[a]DoR is defined as the time from first documented response of CR or PR to the first date of disease progression, start of new therapy, or death, whichever occurs first. CR and PR must have a confirmation scan at least 4 weeks after the initial scan documenting CR or PR. Patients who do not experience disease progression or death, or who receive new cancer therapy, are censored.
CI, confidence interval; cORR, confirmed objective response rate; CR, complete response; DoR, duration of response; HNSCC, head and neck squamous cell carcinoma; N/A, not applicable; NE, not estimable; non-OPSCC, non-oropharyngeal squamous cell carcinoma; OPSCC, oropharyngeal squamous cell carcinoma; PFS, progression-free survival; PR, partial response; Q3W, every 3 weeks.

CONCLUSIONS

- The results presented are in line with the previously reported safety profile of CRB-701,[8] demonstrating that ocular toxicities are the primary TRAEs.
 - Ocular toxicities were largely manageable and reversible through dose delays and reductions as well as prophylactic measures.
- CRB-701 3.6 mg/kg Q3W demonstrated selective antitumor activity with durable responses in heavily pretreated patients with OPSCC whose disease has progressed on at least one line of standard-of-care therapy.
 - Anatomical subtype and HPV status appeared to be the major baseline characteristics leading to a selective response.
- An ongoing dose expansion phase is evaluating the efficacy and safety of CRB-701 in combination with pembrolizumab in the first-line setting for OPSCC.
- Collectively, these findings demonstrate the potential of CRB-701 as a therapeutic option for second- and third-line treatment of patients with OPSCC with high unmet medical need, and support the continued development of CRB-701 in a pivotal study.

References

1. Barsouk A *et al. Med Sci (Basel)* 2023;11:42.
2. Lim YX, D'Silva NJ. *Oncogene* 2024;43:543–54.
3. Fakhry C *et al. Cancer* 2018;124:2125–33.
4. Sanders C *et al. Oncotarget* 2022;13:1166–73.
5. Park JC *et al. Oncologist* 2025;30:oyaf043.
6. Wang J *et al. Sci Rep* 2019;9:13404.
7. Xu S *et al. Front Immunol* 2022;12:798424.
8. Perez C *et al.* European Society for Medical Oncology Congress, 2025. Poster.
9. Astellas Pharma. Summary of product characteristics, enfortumab vedotin. EMA, 2024. https://www.ema.europa.eu/en/documents/product-information/padcev-epar-product-information_en.pdf (Accessed May 5, 2026).
10. Zhang J *et al. Ann Oncol* 2025;36:934–43.
11. Swiecicki PL *et al. J Clin Oncol* 2025;43:578–88.
12. Powles T *et al. N Engl J Med* 2021;384:1125–35.
13. Eisenhauer EA *et al. Eur J Cancer* 2009;45:228–47.

Acknowledgments

Medical writing support was provided by Katy Sutcliffe PhD of PharmaGenesis Cardiff, Cardiff, UK, and funded by Corbus Pharmaceuticals, Inc., in accordance with Good Publication Practice (GPP 2022) guidelines (www.ismpp.org/gpp-2022).

Funding

Study sponsored by Corbus Pharmaceuticals, Inc.

Presented at the American Society of Clinical Oncology (ASCO) 2026 Annual Meeting, May 29–June 2, 2026, Chicago, IL, USA